1100

Comparative in vitro activity of XRP 2868, a new, oral streptogramin

D. Felmingham[1], M.J. Robbins[1], J.Shackcloth[1], C.Dencer[1], L. Williams[1], C. Couturier[2] and A. Bryskier[2]

[1]G.R. Micro Ltd, 7-9 William Road, London NW1 3ER UK; [2]Sanofi Aventis, Paris cedex 75012, France

D. Felmingham
G.R. Micro Ltd
7–9 William Road
London
NW1 3ER, UK

Tel: +44 (0) 20 7388 7320
Fax: +44 (0) 20 7388 7324
Email: d.felmingham@grmicro.co.uk

INTRODUCTION

XPR 2868 is an investigational, oral streptogramin comprised of a 70:30 ratio mixture of RPR 132552 (a pristinamycin IIB derivative) and RPR 202868 (a pristinamycin IA derivative) (Figure 1).[1]

In this study, the in vitro activity of XRP 2868 was determined against pathogenic bacterial isolates causing respiratory tract, skin and soft tissue, and genito-urinary tract infections, and compared with that of its constituents (RPR 132552 and RPR 202868), pristinamycin, quinupristin/dalfopristin (Synercid®) and erythromycin A.

Figure 1. Chemical structures of RPR 132552 and RPR 202868.

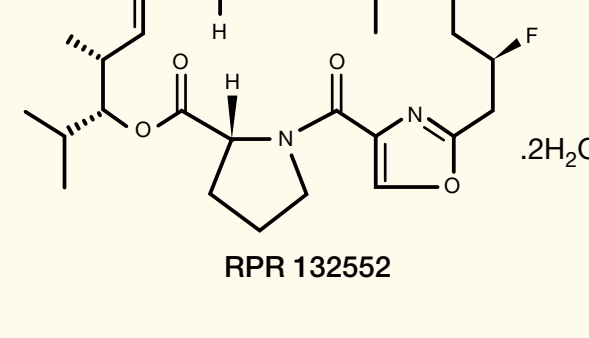

RPR 132552

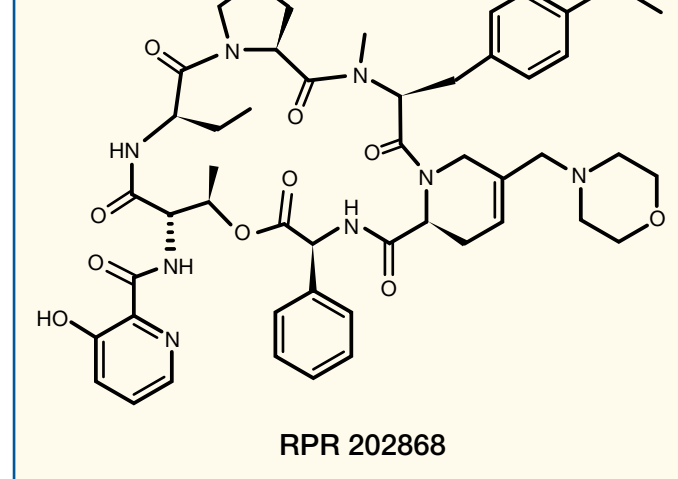

RPR 202868

MATERIALS AND METHODS

Bacterial isolates

Bacterial isolates used in this study were cultured from clinical material collected recently from patients with respiratory tract, skin and soft tissue, and genito-urinary tract infections.

Antimicrobials

Antimicrobial compounds were provided by Sanofi Aventis, Paris, France.

Determination of minimum inhibitory concentrations (MIC)

MIC of XRP 2868 and the comparator compounds, for the majority of isolates examined in this study, were determined using appropriate CLSI methods.[2]

MIC for isolates of *Chlamydophila* (Chlamydia) *pneumoniae* and Chlamydia trachomatis were determined using a tissue culture incorporation method in McCoy cells, – for isolates of *Legionella pneumophila* using an agar incorporation method in buffered yeast extract agar supplemented with lysed horse blood (5% w/v final concentration) and, – for isolates of *Mycoplasma* spp. and *Ureaplasma urealyticum* using a broth microdilution method.[3]

RESULTS

Streptococcus pneumoniae *(Tables 1 and 2)*

XRP 2868 was highly active (MIC_{50}, 0.25 mg/L; MIC_{90} 0.5 mg/L; MIC range, 0.06–2 mg/L) against isolates of *S. pneumoniae*, including those strains resistant to erythromycin (including *erm* B, *mef* A, *erm* B + *mef* A, *erm* A [sub-class *erm* TR] and ribosomal mutation genotypes), telithromycin, levofloxacin and penicillin. Generally, XRP 2868 was four times more active than quinupristin/dalfopristin (Synercid®) and twice as active as pristinamycin, against this species.

Table 1. Comparative in vitro activity of XRP 2868 (mg/L), and its constituents, against *Streptococcus pneumoniae* (n=106).

Streptococcus pneumoniae – phenotype/genotype (n)	XRP 2868			RPR 132552			RPR 202868			quinuspristin/dalfopristin			pristinamycin			erythromycin A		
	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range
– pen^S isolates (11)	–	–	0.12–0.25	–	–	2–8	–	–	2–4	–	–	0.25–0.5	–	–	0.12–0.5	–	–	0.03–0.06
– pen^I isolates (12)	–	–	0.12–0.5	–	–	2–8	–	–	4–64	–	–	0.25–1	–	–	0.12–1	–	–	0.03–>32
– pen^R isolates (13)	–	–	0.06–0.5	–	–	0.25–8	–	–	4–64	–	–	0.25–2	–	–	0.12–1	–	–	0.06–>32
– ery^S isolates (14)	–	–	0.12–0.25	–	–	1–8	–	–	2–4	–	–	0.25–1	–	–	0.12–0.5	–	–	0.03–0.06
– ery^R isolate genotypes																		
– *erm* B (10)	–	–	0.12–0.5	–	–	1–8	–	–	8–32	–	–	0.5–4	–	–	0.25–1	–	–	all >32
– *mef* A (9)	–	–	0.06–0.25	–	–	1–8	–	–	2–4	–	–	0.25–1	–	–	0.12–1	–	–	2–16
– *erm* B + *mef* A (9)	–	–	0.12–0.5	–	–	2–8	–	–	4–32	–	–	0.5–4	–	–	0.25–1	–	–	4–>32
– *erm* A (*erm* TR) (3)	–	–	0.12–0.25	–	–	0.5–4	–	–	2–4	–	–	0.5–1	–	–	0.25–0.5	–	–	0.5–1
– ribosomal mutants (20)	–	–	0.25–2	–	–	0.5–64	–	–	4–>64	–	–	0.5–8	–	–	0.5–8	–	–	1–>32
– telithromycinR isolates (10)	–	–	0.12–0.5	–	–	0.5–4	–	–	16–>64	–	–	1–4	–	–	0.5–2	–	–	all >32
– levofloxacinR isolates (9)	–	–	0.12–0.25	–	–	0.5–4	–	–	2–4	–	–	0.5–1	–	–	0.12–0.5	–	–	0.06–>32
All isolates (106)	**0.25**	**0.5**	**0.06–2**	**4**	**8**	**0.25–64**	**8**	**64**	**2–>64**	**1**	**4**	**0.25–8**	**0.5**	**1**	**0.12–8**	**>32**	**>32**	**0.03–>32**

Table 2. Line listing of the comparative in vitro activity of XRP 2868, and its constituents, against 20 erythromycin A -resistant isolates of *Streptococcus pneumoniae* with characterised ribosomal mutations.

Isolate no	Ribosomal mutations			MIC (mg/L)					
	23SrRNA (no. of copies)	L4 riboprotein	L22 riboprotein	XRP 2868	RPR 132552	RPR 202868	quinupristin/ dalfopristin	pristinamycin	erythromycin A
P 1024019	A2059G(2)	W	W	0.25	4	4	1	0.5	16
P 1521024	A2059G(3)	W	W	0.25	16	8	2	0.5	32
P 1526033	A2058G(2)	W	W	0.25	8	8	2	1	>64
P 2079029	A2058G(3)	W	W	0.25	4	8	1	0.5	>64
PU 1044023	A2058G(4)	W	W	0.25	8	16	1	0.5	>64
PU 1037089	A2058U(3)	I59V	W	0.25	8	32	2	1	>64
PU 1161006	A2059G(1),A2059C(1)	W	W	0.25	0.5	16	1	0.5	>64
PU 1004017	A2059G(3), G2057A(4)	W	W	0.5	8	16	4	2	>64
PU 1049283	C2611A(4)	W	W	0.5	4	16	2	1	1
P 2026064	W	W	109RTAHIT114 (tandem duplication)	2	4	16	8	2	2
P 1080019	A2059G(4)	W	G95D	1	8	32	4	1	16
P 2523249	W	K68S, 69GTGR72 (12bp deletion)	W	1	16	16	4	2	0.25
B 1021014	W	68KG69–68KEG69 (3bp insertion)	W	1	8	8	4	1	0.5
PU 1071099	W	64PWRQ67–64P_Q67 (6bp deletion)	W	0.25	4	4	0.5	0.5	1
PU 1011003	W	69GT70–69VP70	W	1	16	32	4	2	4
PU 1041009	W	69GTG71–69TPS71, V88I	W	0.5	64	8	2	1	>64
PU 1177040	W	E30K, 69GTG71–69TPS71, V88I	W	0.5	64	8	1	1	>64
P 3026056	W	K68Q	W	0.5	4	>128	2	1	>64
P 1008006	C2611G(3)	W	W	2	4	>128	8	8	32
P 1024010	A2059G(4)	W	W	0.25	4	4	1	0.5	32

Staphylococcus aureus *and* Streptococcus pyogenes *(Table 3)*

XRP 2868 was highly active (MIC_{50} / MIC_{90} ,0.25 mg/L; MIC range, 0.06–0.5 mg/L) against both methicillin–susceptible and methicillin-resistant isolates of *S. aureus* including those strains resistant to the macrolides (erythromycin A). Generally, XRP 2868 was 2–4 times more active than quinupristin/dalfopristin (Synercid®) and pristinamycin, against this species.

Table 3. Comparative in vitro activity of XRP 2868 (mg/L), and its constituents, against *Staphylococcus aureus*, *Streptococcus pyogenes* and other Gram positive species.

Species – phenotype/genotype (n)	XRP 2868			RPR 132552			RPR 202868			quinuspristin/dalfopristin			pristinamycin			erythromycin A		
	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range
S. aureus – MSSA (41)	0.12	0.25	0.06–0.25	0.5	0.5	0.12–1	16	32	8–≥128	0.5	0.5	0.25–1	0.25	0.25	0.12–0.5	0.25	≥128	0.25–≥128
S. aureus – MRSA (53)	0.25	0.25	0.06–0.5	0.5	0.5	0.12–1	≥128	≥128	16–≥128	0.5	1	0.12–1	0.5	1	0.12–1	≥128	≥128	0.25–≥128
S. pyogenes – ery^S (5)	–	–	0.03–0.06	–	–	0.06–0.12	–	–	all 16	–	–	0.25–0.5	–	–	0.06–0.12	–	–	0.03–0.06
S. pyogenes – *erm*B (35)	–	–	0.06–0.12	–	–	0.06–0.12	–	–	8–≥128	–	–	0.25–1	–	–	0.12–0.25	–	–	1–≥128
S. pyogenes – *erm*A (TR) (12)	–	–	all 0.06	–	–	0.06–0.12	–	–	16–64	–	–	all 0.25	–	–	0.06–0.12	–	–	2–≥128
S. pyogenes – *mef* A (8)	–	–	all 0.06	–	–	0.06–0.12	–	–	8–16	–	–	all 0.25	–	–	0.06–0.12	–	–	all 16
Streptococcus spp. (63)	0.12	0.5	0.03–2	0.5	32	0.06–64	16	32	8–64	1	4	0.25–8	0.25	0.5	0.06–2	0.06	0.12	0.03–>64
E. faecalis (36)	1	4	0.06–4	>64	>64	0.25–>64	128	>128	2–>128	8	16	0.5–32	2	8	0.12–16	>64	>64	0.25–>64
E. faecium (38)	0.25	0.5	0.06–2	1	64	0.25–>64	128	>128	2–>128	0.5	2	0.25–4	0.25	0.5	0.12–2	>64	>64	0.12–>64
L. monocytogenes (23)	0.5	0.5	0.25–0.5	32	32	16–32	4	8	4–8	1	1	0.5–1	0.5	0.5	0.25–0.5	0.12	0.12	0.12–0.5
C. jeikeium (30)	0.25	1	0.03–2	0.5	1	0.03–1	32	64	32–64	0.5	2	0.25–2	0.25	2	0.06–4	>64	>64	0.03–64
A. haemolyticum (9)	–	–	≤0.015–0.03	–	–	≤0.015–0.03	–	–	all 16	–	–	0.12–0.25	–	–	≤0.03–0.06	–	–	≤0.015–0.03

XRP 2868 was highly active (MIC_{50} / MIC_{90}, 0.06 mg/L; MIC range, 0.03–0.12 mg/L) against isolates of *S. pyogenes*, including *erm* B, *erm* A (sub-class *erm* TR) and *mef* A genotypes of macrolide (erythromycin A)- resistant strains. Generally, XRP 2868 was four times more active than quinupristin/dalfopristin (Synercid®) and twice as active as pristinamycin against this species.

Other Gram positive species (Table 3)

XRP 2868 demonstrated potent activity against isolates of 'viridans' *Streptococcus* spp. *Enterococcus faecium*, *Listeria monocytogenes*, *Corynebacterium jeikeium* and *Arcanobacterium haemolyticum*, with all isolates inhibited by 2 mg/L, or less, and MIC_{90} in the range 0.03–1mg/L. Isolates of *E. faecalis* were somewhat less susceptible (MIC_{50} 1mg/L: MIC_{90} 4mg/L) than those of *E. faecalis*. For all species, XRP 2868 retained high activity against macrolide (erythromycin A)- resistant strains and was generally 2–8 times more active than the combination of quinupristin/dalfopristin (Synercid®).

Haemophilus *spp.,* Moraxella catarrhalis*,* Bordetella pertussis *and* Neisseria *spp. (Table 4)*

The in vitro activity of XRP 2868 against isolates of *Haemophilus influenzae* was particularly noteworthy (MIC_{50}, 0.5 mg/L; MIC_{90} 1 mg/L; MIC range, 0.12–2mg/L) being 8 times greater than that of quinupristin/dalfopristin (Synercid®) and 4 times greater than that of pristinamycin. This level of activity embraced strains which were either β-lactamase producing or non-producing, BLNAR, ciprofloxacin-resistant and erythromycin A-resistant.

XRP 2868 was 4–8 fold less active against isolates of *H. parainfluenzae* (MIC_{50} 1 mg/L: MIC_{90} 8 mg/L) than against those of *H. influenzae*.

XRP 2868 was highly active against isolates of *M. catarrhalis*, *B. pertussis*, *N. menigitidis*, and *N. gonorrhoeae* with MIC_{90} in the range 0.06–0.25 mg/L. This activity was generally 4–8 times greater than that of quinupristin /dalfopristin (Synercid®).

Table 4. Comparative in vitro activity of XRP 2868 (mg/L), and its constituents, against *Haemophilus* species, *Moraxella catarrhalis*, *Bordetella pertussis* and *Neisseria* species.

Species – phenotype/genotype (n)	XRP 2868			RPR 132552			RPR 202868			quinuspristin/dalfopristin			pristinamycin			erythromycin A		
	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range
H. influenzae																		
– β–lactamase–ve (19)	–	–	0.25–2	–	–	0.12–2	–	–	all ≥128	–	–	2–16	–	–	1–8	–	–	4–16
– β–lactamase+ve (18)	–	–	0.12–1	–	–	0.12–1	–	–	all ≥128	–	–	2–8	–	–	0.5–8	–	–	1–8
– BLNAR*(5)	–	–	0.5–1	–	–	0.25–1	–	–	all ≥128	–	–	4–8	–	–	2–4	–	–	2–4
– ciprofloxacin^R (3)	–	–	0.5–1	–	–	0.25–1	–	–	all ≥128	–	–	8–16	–	–	2–4	–	–	2–16
– erythromycin^R (2)	–	–	0.25/2	–	–	0.12/2	–	–	both ≥128	–	–	8/16	–	–	1/4	–	–	both ≥128
– all isolates (52)	0.5	1	0.12–2	0.25	1	0.12–2	≥128	≥128	all ≥128	4	8	2–16	2	4	0.5–8	4	8	1–≥128
H. parainfluenzae (20)	2	8	0.25–8	2	4	0.25–4	>64	>64	all >64	32	32	4–64	8	16	1–16	2	8	1–16
M. catarrhalis (30)	0.06	0.12	≤0.015–0.12	0.12	0.12	≤0.015–0.12	≥128	≥128	16–≥128	1	1	0.25–2	0.25	0.5	0.06–0.5	0.12	0.12	0.03–0.25
B. pertussis (10)	–	–	0.03–0.06	–	–	0.03–0.06	–	–	16–32	–	–	0.12–0.25	–	–	all 0.06	–	–	0.06–0.12
N. meningitidis (20)	0.03	0.06	≤0.015–0.06	0.03	0.03	≤0.015–0.06	32	>64	8–>64	0.25	0.5	0.06–1	0.06	0.12	0.03–0.25	0.12	0.25	0.03–0.25
N. gonorrhoeae (30)	0.12	0.25	0.03–0.5	0.12	0.25	0.06–0.25	>64	>64	all >64	0.5	1	0.12–4	0.12	0.5	0.06–1	1	2	0.25–4

*BLNAR = β–lactamase negative, ampicillin – resistant (MIC = ≥4mg/L)

Mycoplasma spp., Ureaplasma urealyticum, Legionella *spp.,* Chlamydophila *(Chlamydia)* pneumoniae *and* Chlamydia trachomatis *(Table 5).*

XRP 2868 was highly active against isolates of these species with MIC_{90} in the range 0.06–0.5mg/L and all isolates inhibited by 0.5mg/L, or less. This activity was generally 4–8 times greater than that of the combination of quinupristin/dalfopristin (Synercid®).

Table 5. Comparative in vitro activity of XRP 2868 (mg/L), and its constituents, against *Mycoplasma* species, *Ureaplasma urealyticum*, *Legionella* species, *Chlamydophila* (Chlamydia) *pneumoniae* and *Chlamydia trachomatis*.

Species – phenotype/genotype (n)	XRP 2868			RPR 132552			RPR 202868			quinuspristin/dalfopristin			pristinamycin			erythromycin A		
	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range	MIC_{50}	MIC_{90}	range
Mycoplasma pneumoniae (20)	0.12	0.25	0.03–0.25	0.12	0.5	0.06–0.5	≥64	≥128	16–≥128	0.12	0.25	0.03–0.5	0.25	0.5	0.12–0.5	0.015	0.03	0.004–0.03
Mycoplasma hominis (20)	0.25	0.25	0.12–0.5	0.5	0.5	0.12–0.5	32	>128	16–>128	1	2	0.25–4	0.5	0.5	0.25–2	>128	>128	all ≥128
Ureaplasma urealyticum (19)	0.25	0.5	0.12–0.5	0.5	2	0.25–8	128	>128	16–>128	1	2	0.5–2	0.5	2	0.25–2	0.25	0.5	0.12–1
Legionella spp.																		
– *pneumophila* (16)	–	–	0.015–0.06	–	–	0.03–0.12	–	–	0.5–4	–	–	0.25–1	–	–	0.03–0.12	–	–	0.12–1
– *dumoffii* (1)	–	–	0.06	–	–	0.12	–	–	0.5	–	–	0.25	–	–	0.06	–	–	0.12
– *longbeachae* (1)	–	–	0.03	–	–	0.06	–	–	0.5	–	–	0.25	–	–	0.06	–	–	0.5
– *micdadei* (1)	–	–	0.03	–	–	0.06	–	–	0.5	–	–	0.5	–	–	0.06	–	–	0.5
– *bozemanii* (1)	–	–	0.03	–	–	0.06	–	–	0.5	–	–	0.25	–	–	0.06	–	–	0.25
– all isolates (20)	0.03	0.06	0.015–0.06	0.06	0.12	0.03–0.12	0.5	2	0.5–4	0.5	0.5	0.25–1	0.06	0.12	0.03–0.12	0.25	0.5	0.12–1
Chlamydophila (Chlamydia) *pneumoniae* (5)	–	–	all 0.25	–	–	all 0.5	–	–	all ≥128	–	–	2–4	–	–	0.5–1	–	–	1–2
Chlamydia trachomatis (10)	–	–	0.12–0.25	–	–	0.06–0.25	–	–	all ≥128	–	–	0.5–1	–	–	0.12–0.25	–	–	0.12–0.5

CONCLUSION

XRP 2868 is a suitable candidate for further investigation as a potential, oral anti-infective therapeutic agent.

ACKNOWLEDGEMENT

This work was financially supported by Sanofi Aventis, Paris, France.

REFERENCES

1 Pankuch et al. (2003) Activities of a new oral streptogramin, XRP 2868, compared to those of other agents against *Streptococcus pneumoniae* and *Haemophilus species*. *Antimicrobial Agents and Chemotherapy* 47 (10): 3270–3274.

2 NCCLS. (2003) Methods for dilution antimicrobial susceptibility tests for bacteria that grow Aerobically; Approved Standard – Sixth Edition. *NCCLS Document M7-A6 (ISBN 1-56238-486-4)* NCCLS, Wayne, PA 19087, USA. *(NB. Now renamed CLSI [Clinical Laboratory Standards Institute]).*

3 Ridgway et al. (1997) The in vitro activity of grepafloxacin against *Chlamydia* spp., *Mycoplasma* spp., *Ureaplasma urealyticum* and *Legionella* spp. *Journal of Antimicrobial Chemotherapy* 40 (Suppl A): 31–34.