F-2030

Comparative Prevalence in European Union - Evaluation of the *in vitro* Activity of NXL103 (Formerly XRP2868) and Macrolides Against Common Respiratory Pathogens.

novexel
novel therapies for infectious disease
Tél: 01 57 14 07 77 Fax: 01 48 46 39 26
E-mail: info@novexel.com
Contact: John Lowther
Parc Biocitech - 102, route de Noisy
F- 93230 Romainville - France

A. Bryskier[2], J. Lowther[1], J. Hodgson[1] and the European Study Group*.

1 - Novexel SA, Romainville, France. 2 - Sanofi Aventis, Paris, France.

ABSTRACT

Background: The emergence and spread of macrolide resistance among streptococci involved in respiratory tract infections has reached alarming proportions in some geographic areas.

The research for new antibiotics is essential to overcome this threat in the community and in the hospital setting.

An epidemiological surveillance programme before full development is useful to investigate the potential of a new therapeutic entity.

Methods: A multinational epidemiological survey covering 15 E-U countries was carried out during the winter of 2003-2004 for respiratory pathogens including *S. pneumoniae, S. pyogenes,* and *H. influenzae*. 127 centres were enrolled in this study and the clinical isolates were tested for their susceptibility to NXL103 a new oral streptogramin, and erythromycin A.

Results: MICs for NXL103 and erythromycin A are reported in Table 1.

TABLE 1

Organism	No. tested	MIC (µg/ml) NXL103 50%	NXL103 90%	NXL103 range	Erythromycin A 50%	Erythromycin A 90%	Erythromycin A range
S. pneumoniae	2687	0.25	0.25	0.016-1.0	0.06	64	0.016->64
S. pyogenes	1509	0.03	0.06	0.016-0.5	0.06	8.0	0.016->64
H. influenzae	2418	0.5	1.0	0.03- 4.0	4.0	8.0	0.016->64

The genetic mechanism was investigated for gram-positive cocci resistant to erythromycin A and the correlation with the susceptibility of NXL103 is listed in Table 2.

TABLE 2

Organism	No. tested*	Modal MIC NXL103 (µg/ml) *erm*B	*mef*E	*erm*TR	*mef*A
S. pneumoniae	659	0.25	0.25	0.25	-
S. pyogenes	184	0.03	-	0.03	0.03

*number of isolates resistant to erythromycin A (MIC 1 µg/ml)

Conclusion: NXL103 is active against *S. pneumoniae, S. pyogenes, and H. influenzae* including strains susceptible or resistant to erythromycin A. No isolate resistant to NXL103 has been detected among the 6614 clinical isolates collected. Clinical trials are warranted.

INTRODUCTION

Of the main common pathogens involved in lower (LRTI) and upper respiratory tract infections (URTI), three pathogens are most commonly isolated: *S. pneumoniae, S. pyogenes,* and *H. influenzae. S. pneumoniae and S. pyogenes* have progressively become highly resistant to erythromycin A, and *H. influenzae* has become resistant to ampicillin by β-lactamase production.
During the winter period of 2003-2004, an epidemiological surveillance programme was initiated in 15 European Union countries investigate the susceptibility of these pathogens to a new oral streptogramin: NXL103 (formerly XRP 2868).
NXL103 is a new semisynthetic oral streptogramin consisting of a 30:70 mixture of PI and PII components. PI is characterised by a morpholine ring, and PII by a fluorine substitution (Fig 2).

MATERIALS AND METHODS

The epidemiological programme was divided into (1) a cross validation study between all laboratories performing MICs (2) the evaluation of susceptibility of the collected clinical isolates.

1. Fourteen clinical laboratories were in charge of determining MICs. Each centre interfaced with several local laboratories in their geographic area, and was in charge of collecting and sending the micro-organisms to one of the regional laboratories. One hundred and twenty seven (127) collecting laboratories were involved.

2. MIC determinations were performed by a microdilution method (Trek Diagnostic Systems United Kingdom) following CLSI recommendations. MICs were determined in triplicate for three consecutive days (nine determinations).

RESULTS

1. *S. pneumoniae*
Two thousand six hundred and eighty seven (2687) *S. pneumoniae* isolates were collected from blood and respiratory tracts. Globally, 77.8% were susceptible to penicillin G (resistance rates ranging from 2% in Norway to 50.5% in France). For erythromycin A, for all the countries, the susceptibility was 74.4% (resistance rates ranging from 8% in Sweden to 53.1% in France). The overall resistance to tetracycline was 21.4%. MIC distribution for NXL103 was 0.01 to 1.0 µg/ml (MIC_{50}: 0.25 µg/ml and MIC_{90}: 0.25 µg/ml) (Table 1).

2. *S. pyogenes*
One thousand five hundred and nine (1509) clinical isolates were collected of which 81.6% were from the respiratory tract. All the isolates were susceptible to penicillin G. The lowest prevalence rate for erythromycin A susceptibility was found in Spain (68.2%) and the highest in Sweden and Finland (100%). The overall prevalence rate of tetracycline resistance was 13.8%.
NXL103 MICs ranged from 0.01 to 0.5 µg/ml with MIC_{50}: 0.03 µg/ml and MIC_{90}: 0.06 µg/ml (Table 1).

TABLE 3: NXL103 *in vitro* activity per country (MIC50 and MIC90 µg/ml)

Country	*S. pneumoniae* N*	MIC_{50}	MIC_{90}	*S. pyogenes* N*	MIC_{50}	MIC_{90}	*H. influenzae* N*	MIC_{50}	MIC_{90}
Belgium	93	0.125	0.125	238	0.03	0.03	98	0.25	0.25
Finland	44	0.25	0.25	49	0.03	0.03	53	0.50	0.50
France	495	0.125	0.25	174	0.03	0.03	374	0.50	1.0
Germany	486	0.25	0.25	206	0.03	0.06	426	0.50	0.50
Greece	45	0.25	0.25	52	0.01	0.125	91	0.50	1.0
Italy	229	0.125	0.125	201	0.03	0.03	50	0.50	0.50
Norway	50	0.25	0.25	49	0.06	0.125	50	0.50	1.0
Poland	120	0.25	0.25	51	0.03	0.03	119	0.50	1.0
Portugal	104	0.25	0.25	21	0.03	0.03	103	0.50	1.0
Spain	301	0.25	0.25	151	0.03	0.06	300	0.50	1.0
Sweden	100	0.25	0.25	50	0.03	0.03	96	0.50	1.0
The Netherlands**	276	0.25	0.25	101	0.03	0.03	279	0.50	1.0
United Kingdom	344	0.25	0.25	166	0.03	0.03	379	0.50	1.0
Total	**2687**	**0.25**	**0.25**	**1509**	**0.03**	**0.06**	**2418**	**0.50**	**1.0**

* N: number of isolates.
**The Netherlands centre collected: The Netherlands, Denmark and Luxembourg.

3. *H. influenzae*
A total of 2418 clinical isolates were collected of which 92.2% were from the respiratory tract. Among them 13.5% were resistant to ampicillin by β-lactamase production (ranging from 2% in Italy to 24.8% in France) (Figure 1).

FIGURE1: Geographic distribution of β-lactamase producing strains of *H. influenzae* in the European Union

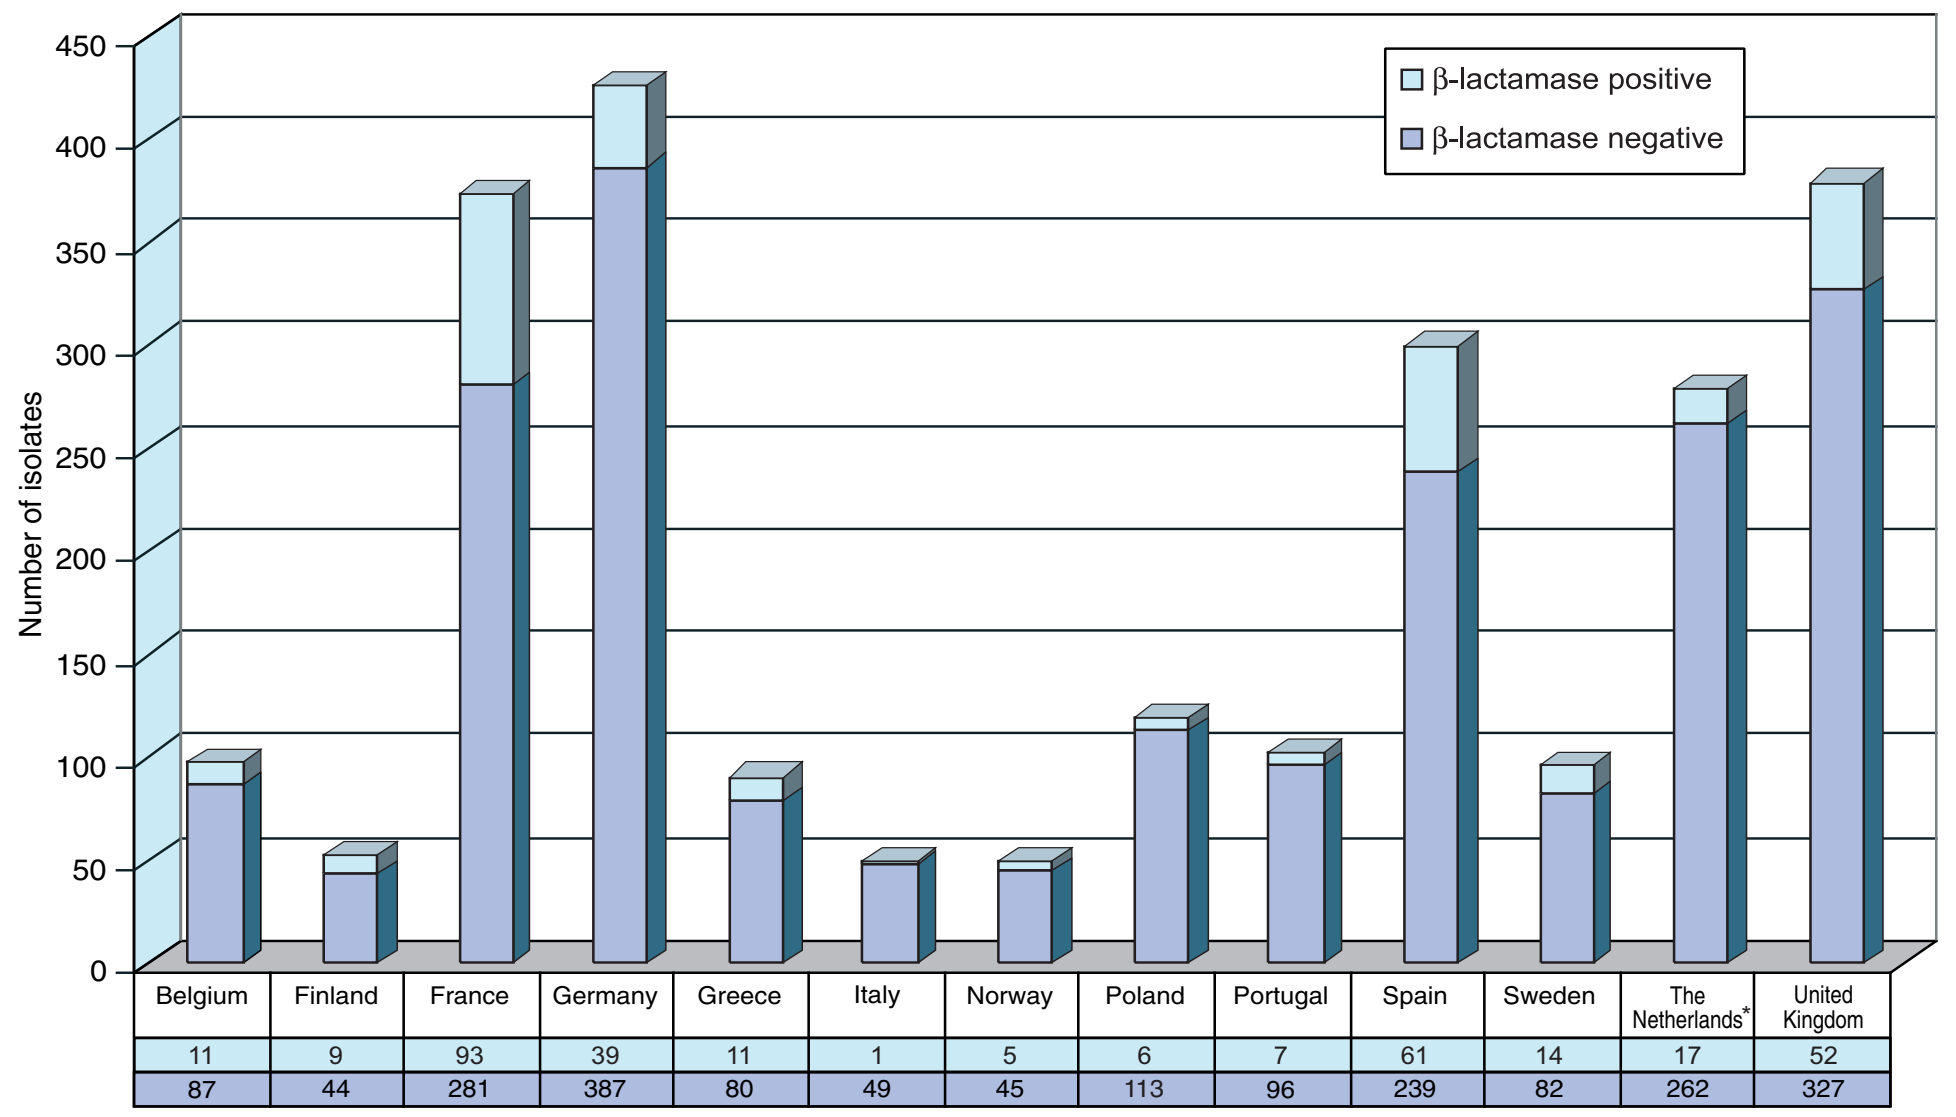

β-lactamase positive
β-lactamase negative
Number of isolates
0
50
100
150
200
250
300
350
400
450
Belgium Finland France Germany Greece Italy Norway Poland Portugal Spain Sweden The Netherlands* United Kingdom
11 9 93 39 11 1 5 6 7 61 14 17 52
87 44 281 387 80 49 45 113 96 239 82 262 327

*The Netherlands includes : Netherlands 5/152 (3.3%), Luxembourg 3/23 (13.0%) and Denmark 9/103 (8.7%)

MICs of NXL103 ranged from 0.03 to 4 µg/ml (Table 1). MIC50 and MIC90 were 0.50 and 1.0 µg/ml respectively.

4.Mechanisms of resistance for *S. pneumoniae* and *S. pyogenes*
All clinical isolates of *S. pneumoniae* and *S. pyogenes* isolates resistant for erythromycin A were investigated for the underlying mechanism of resistance.

For *S. pneumoniae* the most prevalent phenotype of erythromycin A resistance was MLSB (19%) while the M-phenotype was observed in 6% of the isolates.
MLSB and M-phenotypes were observed in about 5% and 8% respectively of *S. pyogenes* isolates.

TABLE 4: Mechanisms of erythromycin A resistance for *S. pneumoniae* and *S. pyogenes*

Genotypes	Phenotypes *S. pyogenes* N	Ery-S	M	MLS_B	*S. pneumoniae* N	Ery-S	M	MLS_B
Erythromycin A-S	1322	1319	3	0	2010	2003	6	1
Erythromycin A-R	(187)				(677)			
mef	79	0	78	1	132	0	130	2
ermB	73	0	10	63	510	0	25	485
ermTR	25	0	20	5	1	0	1	0
ermB + ermTR	1	0	0	1	5	0	0	5
ermB + mef	4	0	0	4	13	0	0	13
ermTR + mef	2	0	2	0	8	0	8	0
No PCR product	3	0	3	0	8	0	3	5
TOTAL	**1509**	**1319**	**116**	**74**	**2687**	**2003**	**173**	**511**

There is a discrepancy between phenotypes and genotypes for 27 out of 677 *S. pneumoniae* isolates (4%) and 16 out of 187 for *S. pyogenes* (8.5%).

For NXL103 whatever the underlying mechanism of resistance to erythromycin A in this survey, MIC values ranged from 0.01 to 0.5 µg/ml and 0.01 to 1.0 µg/ml for *S. pyogenes* and *S. pneumoniae* respectively. (Table 1).

FIGURE 2: Structure

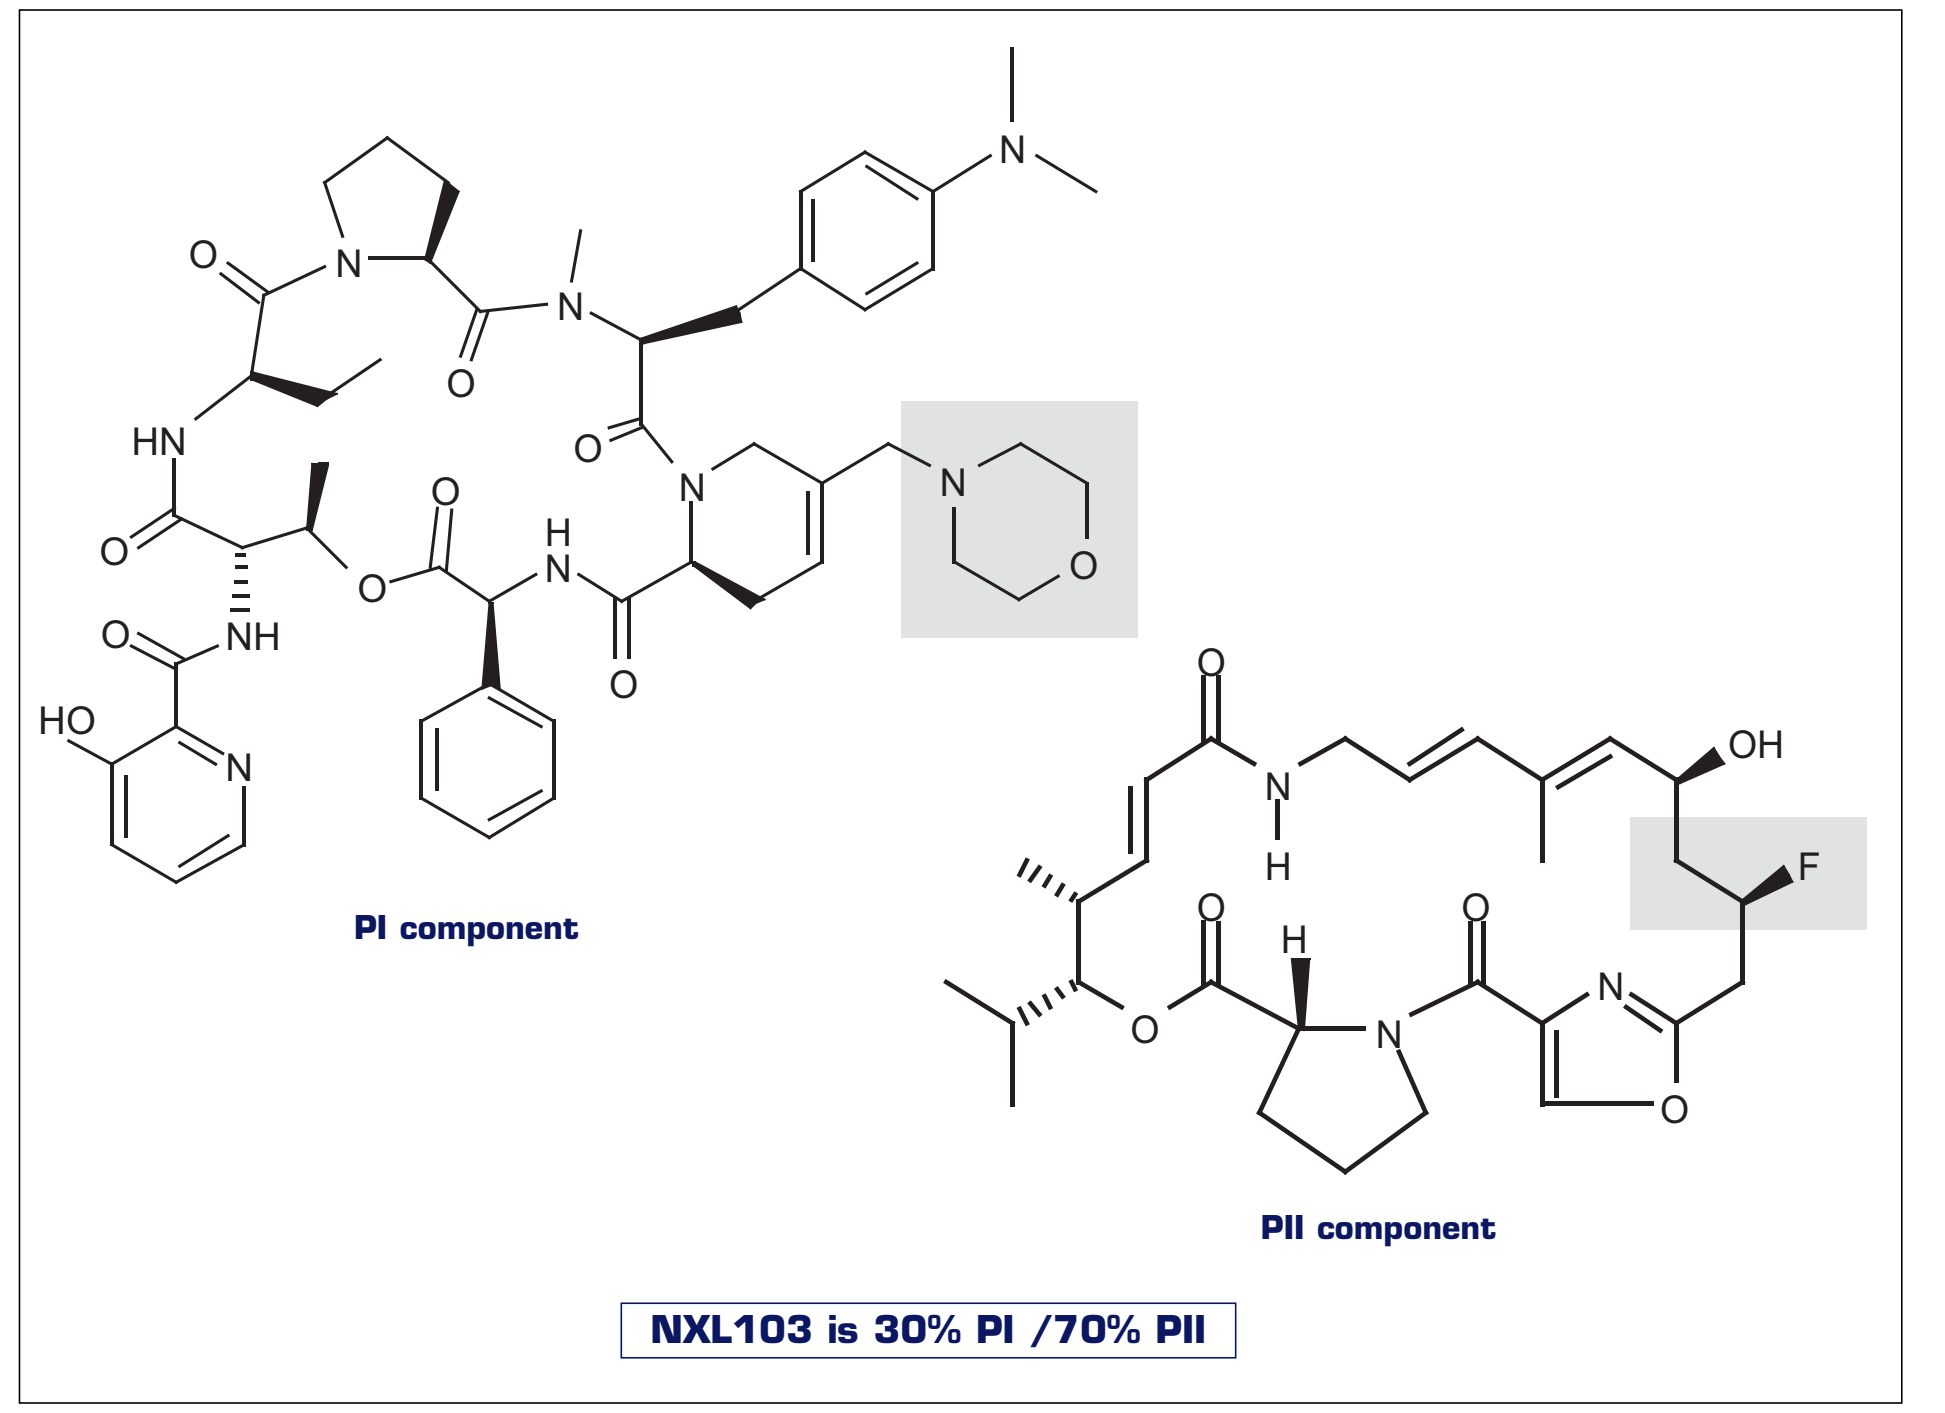

PI component
PII component
NXL103 is 30% PI /70% PII

CONCLUSIONS

- This epidemiological survey throughout the European Union showed that there is a large country-to-country variation in the rates of resistance to penicillin G (*S. pneumoniae*) and erythromycin A (*S. pneumoniae* and *S. pyogenes*);
- For erythromycin A and *S. pneumoniae*, the dominant mechanism of resistance was methylation (erm B). In *S. pyogenes* the methylation and efflux mechanisms were found with similar frequencies.
- NXL103 MICs were similar whatever the underlying mechanism of resistance to erythromycin A.
- *In vitro* activity of NXL103 against *H. influenzae* remained unchanged whatever the susceptibility to ampicillin.
- **It has been demonstrated in a previous study that NXL103 is bactericidal (1). NXL103 is highly active against common pathogens currently isolated in the clinics from respiratory tract infections. The data presented here confirm and are consistent with the results found in previous *in vitro* studies (2,3,4).**

References:
1. Drugeon *et al:* Poster Communication F2116 - 43rd ICAAC - Chicago 2003
Role of each component RPR202868 and RPR132552 in the bactericidal synergism of XRP2868 (NXL103) against *Streptococcus pneumoniae, Haemophilus influenzae* and *Staphylococcus aureus.*
2. Felmingham *et al:* Poster communication - P-1555 - 15th ECCMID - Copenhagen 2005
Comparative *in vitro* activity of XRP2868, a new oral streptogramin
3. Appelbaum *et al:* Antimicrobial Agents and Chemotherapy, 2003, 3270–3274 Vol. 47, No. 10
Activities of a New Oral Streptogramin, XRP 2868, Compared to Those of Other Agents against *Streptococcus pneumoniae* and *Haemophilus Species.*
4. Eliopoulos, G *et al:* Antimicrobial Agents and Chemotherapy 2005, 3034–3039 Vol. 49, No. 7
In vitro Activity of an Oral Streptogramin Antimicrobial, XRP2868, against Gram-Positive Bacteria

*European Study group: Principle investigators:

COUNTRY		INVESTIGATOR
BELGIUM	Prof.	Herman GOOSSENS
BELGIUM	Prof.	Jan VERHAEGEN
FINLAND	Prof.	Pentti HUOVINEN
FRANCE	Prof.	Henri DRUGEON
GERMANY	Prof.	René REINERT
GREECE	Prof..	Alkis VATOPOULOS
ITALY	Dr.	Giuseppe CORNAGLIA

COUNTRY		INVESTIGATOR
NETHERLANDS	Prof.	Jan VERHOEF
NORWAY	Dr.	Arnfinn SUNDSFJORD
POLAND	Prof.	Waleria HRYNIEWICZ
PORTUGAL	Prof.	José MELO-CRISTINO
SPAIN	Prof.	Fernando BAQUERO
SWEDEN	Prof.	Barbro OLSSON-LILJEQUIST
THE UNITED KINGDOM	Prof.	David FELMINGHAM

Presented at 45th ICAAC Washington 2005

terraverde