P-1512

NXL103- Oral Streptogramin: A Phase I, Double-blind, Single Escalating oral dose Study to Evaluate Safety, Tolerability and Pharmacokinetics in Healthy adult male volunteers

novexel
novel therapies for infectious disease
info@novexel.com

M. RANGARAJU1, J. REY2, J.HODGSON1
1 Novexel SA, Romainville, France, 2 Sanofi Aventis, Vitry sur Seine, France.

INTRODUCTION

NXL103 is a new oral anti-infective agent of the streptogramin group. It is composed of two components in a 30/70 ratio (w/w) [PI and PII]. The individual components have limited antibacterial activity, but exhibit potent antibacterial activity (including bactericidal activity) when administered together. Streptogramins are members of the MLSB group of antibiotics. They act on ribosomes leading to inhibition of bacterial protein synthesis. Streptogramin antibiotics display a unique mode of action in that a conformational change in bacterial ribosomes is induced by the PII component which favors the binding of the PI component, resulting in a stable tertiary PI-ribosome-PII complex (1).

Streptogramins generally exert bactericidal activity and rarely exhibit cross-resistance with either macrolides or lincosamides due to the fact that the PII component maintains significant association to ribosomes even in strains exhibiting erythromycin A constitutive resistance (MLSB-C). Unlike macrolides and lincosamides, a double mutation is required for resistance development to streptogramins and therefore occurrence of drug resistant strains should be less frequent.

NXL103 has an in vitro spectrum of activity that includes aerobic Gram-positive cocci [including multi-resistant *S.pneumoniæ*, methicillin resistant *S.aureus* (MRSA)] and aerobic Gram-negative bacteria responsible for respiratory tract infections such as *H.influenzae*, *M.catarrhalis*, and atypical pathogens.
Due to its potency against strains resistant to MLSB and other classes of antibiotics, NXL103 could be an alternative to macrolides, ketolides, quinolones, and beta-lactams in the treatment of community acquired respiratory and skin infections.
We report here the first study performed in humans with NXL103. This study was performed to assess the safety, tolerance, and pharmacokinetic (PK) profile of orally administered escalating single doses of NXL103 in healthy male volunteers.

METHODS

• Study design
• The study was conducted in 3 parts.

• The first part (part A) of the study was a randomized, double blind, placebo-controlled study with six escalating oral doses of NXL103 or placebo each to be administered as a single dose to 6 cohorts of 10 subjects. Starting with the lowest dose, each of the subsequent five doses was administered only if the preceding dose was safe and well tolerated.

• The second part (part B) of the study was an open randomized 2 periods crossover design including a washout period of one week. One cohort of 10 subjects received 2 single doses of 500mg NXL103 (in fasting or fed conditions) to assess the effect of food on the bioavailability of NXL103.

• The third part (part C) of the study was an open randomized 2 periods crossover design including a washout period of one week. A total of 10 subjects received 2 single doses of 500mg NXL103 (either as capsules or as an oral solution) to compare the relative bioavailability of the two formulations.

• Subjects
- Healthy male volunteers aged 18 to 40
- Body Mass Index between 18 and 27 kg/m²
- Negative serology: HIV antibody, hepatitis B surface antigen, hepatitis C antibody,
- Negative urine screen for drugs of abuse
-Written informed consent before enrollment in the study

• Safety assessment
Safety and tolerance were assessed on the basis of adverse events, 12-lead ECG recordings and cardiac monitoring, vital signs (blood pressure and heart rate), and clinical laboratory data (hematology, biochemistry and urinalysis).

• Pharmacokinetics assessment
Blood samples for plasma concentrations of NXL103 and its components were collected predose and 0.5, 1, 1.5, 2, 3, 4, 6, 8, 10, 12, 15 and 24 h postdose. Urine samples were collected predose and 0-4 hours, 4-8 hours, 8-12 hours and 12-24 hours post dose. PI, RPR208880 (a metabolite of PI) and PII were quantified in plasma and urine using a validated LC/MS/MS method with limits of quantitation of 5 ng/mL in plasma and 100 ng/mL in urine, respectively.
Pharmacokinetic analysis was performed from data obtained in plasma. Cmax, tmax, AUC(0-t), AUC and t½ were determined by non compartmental analysis for each analyte. Fractional and cumulative urinary excretion were calculated and expressed as percent of the administered dose.

RESULTS

• Subjects
A total of 80 subjects was enrolled. A summary of demographic characteristics is given in Table 1.

• Safety and tolerability
No serious or severe adverse events were reported during the study.
There was no significant variation in Blood pressure or heart rate. No abnormality on ECG recording was observed during the study. No subject had a PR or QRS interval value above the upper limit of normal during the study.
There were no clinically significant changes compared to baseline for any of the laboratory parameters tested.
A total of 16/48 [33.3%] subjects receiving NXL 103 experienced 24 treatment emergent adverse events considered possibly related to the study medication in part A (Table 2). Among the 16 subjects who experienced possibly related AEs, vomiting (mild or moderate) was observed in 7/48 subjects at doses above 500 mg, orthostatic hypotension of mild to moderate intensity was seen in 6/48 subjects at doses above 125 mg (note that subjects should be in a sitting or supine position for up to 4 hours after drug administration), and nausea of mild intensity in 3/48 subjects at 500 mg and above.
Mild dizziness occurred in one subject, during fed state, in part B.
Loose stools of mild intensity in 2 subjects who received NXL 103 as capsules in part C were also considered possibly related to study drug. One of these 2 subjects from part C also experienced two TEAEs (nausea, loose stool) of mild intensity when NXL 103 was given as an oral solution.

•Pharmacokinetics
The Cmax and AUC increased approximately in proportion with dose within the dose range of 125 to 2000 mg (Table 3 and Figure 1). Both PI and PII components of NXL 103 were rapidly absorbed, PI being slightly more rapidly absorbed than PII. At doses of 500, 1000, 1500 and 2000 mg, the proportion of PI and PII components estimated on mean exposure values was approximately comparable to that administered (30 / 70), indicating that the relative bioavailabilities of PI and PII components are comparable (Table 3 and Figure 2).
Terminal half-life ranged between 2 to 3 hours for PI and between 4 to 6 hours for PII. The pharmacokinetics of the three analytes appeared to be dose-independent. Urinary excretion of PI, PII and a metabolite of PI (RPR208880) corresponds to 2 to 7 %, 0.5 to 2 % and 0.1 to 0.7 % of the administered dose, respectively.
Food increased the bioavailability of PI and PII by approximately 20%. The bioavailability of the solution is 1.6 times higher than that of capsules for PI and practically unchanged for PII.

Table 1. Disposition of subjects and demographics

		Part A (n= 60)	Part B (n= 10)	Part C (n= 10)
Age (years) Mean (range)		28.9 (19-40)	26.7 (20-36)	30 (21-35)
Weight (kg) Mean (range)		76.76 (58.90- 95.70)	79.61 (69.30-95.20)	74.88 (61.90-88.30)
Ethnic group	Caucasian	52/60 (86.7 %)	9/10 (90 %)	9/10 (90 %)
	Black	6/60 (10.0 %)	-	-
	Multiracial	2/60 (3.3 %)	1/10 (10 %)	1/10 (10 %)
BMI (kg/m^2) Mean (range)		23.93 (22.95 – 24.91)	24.93 (22-27.8)	24.35 (22.7-27.5)

Table 2. Number of subjects reporting treatment emergent adverse events considered possibly or probably related to study drug - Part A

Body system Class and Medra term	Placebo (n =12)	125 mg (n =8)	250 mg (n =8)	500 mg (n =8)	1000 mg (n =8)	1500 mg (n =8)	2000 mg (n =8)	ALL NXL 103
Total	0	0	1	2	5	3	5	16 (33.3%)
Gastrointestinal disorders			0	2	4	3	4	13
Vomiting					2	2	3	7
Nausea				1		1	1	3
Loose stools					1		1	2
Abdominal Pain				1				1
Flatulence					1			1
Nervous system disorders				1	1	1	1	4
Dizziness				1		1		2
Headache					1		1	2
Vascular disorders			1		3	1	1	6
Orthostatic hypotension			1		3	1	1	6

Note: The numbers in each column cannot be added because a subject may have had more than one adverse event.

Table 3. Mean pharmacokinetic parameters (CV%) of the components of NXL 103- Study Part A

Compound	Dose*(mg)	Tmax (h) (range)	Cmax (ng/mL)	AUC(0-t) (ng.h/mL)
PII	125	2 (0.5 – 3)	171 (35)	509 (47)
	250	1.75 (1.5 – 3)	220 (29)	750 (50)
	500	1.5 (0.5 – 4)	456 (46)	1740 (59)
	1000	2.5 (1 – 4)	1050 (45)	5020 (53)
	1500	3 (1 – 4)	1140 (38)	5180 (36)
	2000	3 (3 – 3)	1770 (26)	8070 (39)
PI	125	1 (0.5 – 2)	38.7 (81)	56.9 (67)
	250	1 (0.5 – 1.5)	93.4 (39)	184 (43)
	500	1 (0.5 – 2)	287 (80)	647 (62)
	1000	2 (1 – 4)	486 (38)	1950 (54)
	1500	2 (1 – 4)	498 (21)	2060 (21)
	2000	3 (2 – 3)	1150 (36)	4210 (27)
Total PI+ PII + M	125	1.75 (0.5 – 3)	203 (29)	576 (46)
	250	1.5 (0.5 – 2)	296 (29)	967 (46)
	500	1.25 (0.5 – 4)	749 (49)	2510 (55)
	1000	2.5 (1 – 4)	1600 (39)	7260 (50)
	1500	2 (1 – 4)	1700 (21)	7560 (23)
	2000	3 (2 – 3)	3040 (20)	13100 (24)

*: vomiting subjects excluded; **: only one value; M = metabolite

Figure 1. Exposure of PI and PII components in relation to dose – Study Part A

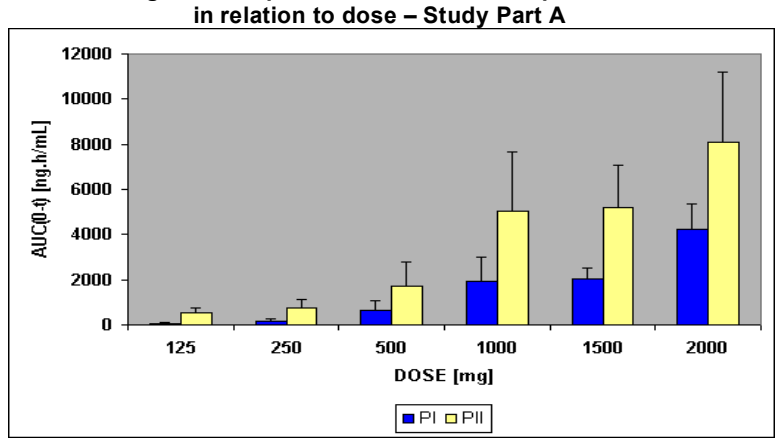
12000
10000
8000
6000
4000
2000
0
AUC(0-t) [ng.h/mL]
125
250
500
1000
1500
2000
DOSE [mg]
PI PII

Figure 2 – Exposure of PI and PII in relation to the ratio of PI (30%)and PII (70%) administered

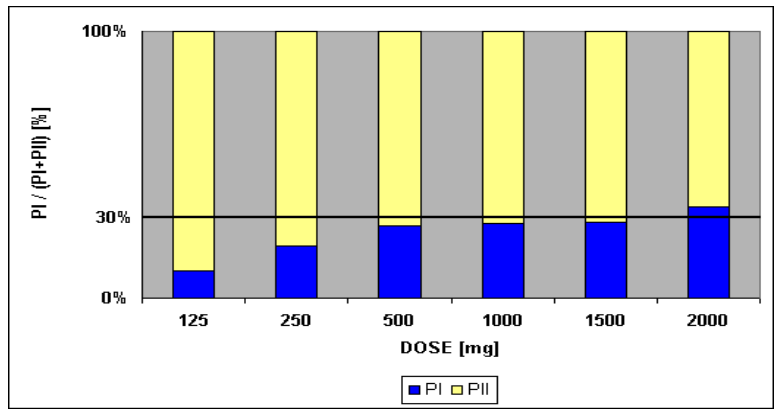
100%
30%
0%
PI / (PI+PII) [%]
125
250
500
1000
1500
2000
DOSE [mg]
PI PII

CONCLUSIONS

NXL103 administered as single doses (125 mg, 250 mg, 500 mg, 1000mg, 1500 mg, 2000 mg) in healthy human volunteers was safe and well tolerated. Pharmacokinetic analyses showed that both PI and PII components of NXL 103 were rapidly absorbed.
The Cmax and AUC increased approximately in proportion with dose within the dose range of 125 to 2000 mg.

Références

1. Cocito C, Di Giambattista M, Nyssen E, and Vannuffel P. : Inhibition of protein synthesis by streptogramins and related antibiotics;J Antimicrob Chemother, 1997 May; 39 Suppl A: 7-13