1348

In vitro activity of AVE1330A, a novel β-lactamase inhibitor, in combination with aztreonam or ceftazidime against ceftazidime-resistant isolates of species of the *Enterobacteriaceae*

J. Shackcloth[1], L. Williams[1], J. Northwood[1], A. Bryskier[2] and D. Felmingham[1]

[1]G.R. Micro Ltd, 7–9 William Road, London NW1 3ER UK; [2]Sanofi Aventis, Paris cedex 75012, France

D. Felmingham
G.R. Micro Ltd
7–9 William Road
London
NW1 3ER, UK

Tel: +44 (0) 20 7388 7320
Fax: +44 (0) 20 7388 7324
Email: d.felmingham@grmicro.co.uk

INTRODUCTION

β-lactam antimicrobials are important antibacterial agents because collectively they offer clinical efficacy against a wide range of pathogenic species and have a very low toxicity when used in humans.[1]

However, the effectiveness of this class of antimicrobials is under considerable threat from the evolution of resistance, particularly that mediated by the Class A extended spectrum β-lactamases (ESBL) and the Class C de-repressed, chromosomally located cephalosporinases.[2,3]

Thus, the therapeutic value of third generation cephalosporins such as ceftazidime, and the monobactam, aztreonam, is now compromised.

As β-lactamases have evolved by mutation and selection to include an increasing number of β-lactams within their hydrolytic spectrum, the pharmaceutical industry has responded by pursuing two research strategies:

1. Structural modification of existing molecules to block β-lactamase active-site binding.
2. The development of β-lactamase inhibitors to protect existing β-lactams from hydrolysis.

Three β-lactamase inhibitors are available, currently, in antibacterial combinations, – clavulanic acid, sulbactam and tazobactam. However, these compounds have a limited spectrum of inhibitory activity confined, generally, to Class A β-lactamases and even within this class inhibitor-resistant enzymes are now recognised. Importantly, none of these β-lactamase inhibitors is effective against strains of species of the *Enterobacteriaceae* producing Class C de-repressed, chromosomally located, cephalosporinases.[4,5,6,7]

AVE1330A is an investigational, non β-lactam, β-lactamase inhibitor (Figure 1).[8]

Figure 1. Chemical structure of AVE1330A

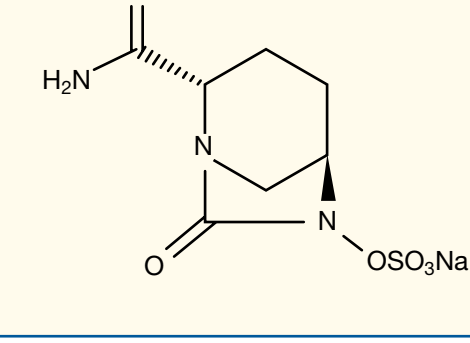

In this study, the ability of AVE1330A to protect ceftazidime and aztreonam against hydrolysis by a variety of β-lactamases, was investigated.

MATERIALS AND METHODS

Bacterial isolates

Bacterial isolates used in this study were derived from clinical material examined recently by colleagues from hospitals and reference centres worldwide. Isolates were maintained with minimal sub-culture, deep-frozen at –70°C as a dense suspension in a high protein matrix of undiluted horse serum.

Antimicrobials

The following antimicrobials were used: AVE1330A (Lot no 92985, Aventis Pharma), amoxycillin (Lot no. 112K0481, Sigma), clavulanic acid (Lot no. BRL 14151, GlaxoSmithKline), aztreonam (Lot no. 6016E, ICN Biomedical), cefoxitin (Lot no. 2251042, Sigma) and ceftazidime (Lot no. 13K0525, Sigma).

Determination of minimum inhibitory concentrations (MIC)

MIC were determined using methods for antimicrobial susceptibility testing published by the Clinical Laboratory Standards Institute (CLSI), formerly the NCCLS.[9]

RESULTS AND DISCUSSION

The results of this study are presented in Tables 1-3.

Ceftazidime-resistant strains of species of the Enterobacteriaceae *(Table 1)*

A total of 45 ceftazidime-resistant strains of species of the *Enterobacteriaceae* were examined (*Escherichia coli* (12), *Citrobacter freundii* (6), *Klebsiella pneumoniae* (11), *Klebsiella oxytoca* (1), *Enterobacter cloacae* (13) and *Morganella morganii* (2)).

The addition of AVE1330A, at a fixed concentration of 4 mg/L, to ceftazidime and aztreonam was highly effective in protecting each antimicrobial against the hydrolytic activity of β-lactamases produced by ceftazidime-resistant strains of *Escherichia coli.* MIC of ceftazidime and aztreonam in combination with AVE1330A were in the range ≤0.03–1 mg/L compared to 16–>64 mg/L for ceftazidime, alone, and, ≤0.06–0.25 mg/L compared to 4–>64 mg/L for aztreonam, alone.

A similar picture was seen with strains of *Citrobacter freundii* (MIC range: ceftazidime = 32–>128 mg/L; ceftazidime + AVE1330A = 0.25–1 mg/L: aztreonam = 4–64 mg/L; aztreonam + AVE1330A = 0.12–0.25 mg/L).

Eleven (11) strains of ceftazidime – resistant *Klebsiella pneumoniae* were highly susceptible to the combination of ceftazidime + AVE1330A (MIC range: ≤0.03–1 mg/L) and to that of aztreonam + AVE1330A (MIC range: ≤0.06–0.5 mg/L).

The activity of aztreonam + AVE1330A (fixed concentration, 4 mg/L) against a single strain of *Klebsiella oxytoca* (MIC: 0.25 mg/L) was 16-fold greater that of the combination of AVE1330A with ceftazidime (MIC: 4 mg/L), although both combinations were considerably more active than either aztreonam or ceftazidime, alone.

The combinations of both aztreonam and ceftazidime, with a fixed concentration of 4 mg/L AVE1330A, were both highly effective against the majority of the 13 strains of *Enterobacter cloacae* tested with MIC in the range of ≤0.06–1 mg/L and 0.5–8 mg/L, respectively. One (1) strain required an MIC of 2 mg/L ceftazidime + AVE1330A compared to 1 mg/L aztreonam + AVE1330A and one (1) required an MIC of 8 mg/L ceftazidime + AVE1330A compared to 0.5 mg/L aztreonam + AVE1330A. Two (2) strains of ceftazidime-resistant *Morganella morganii* were both highly susceptible to the combination of either aztreonam or ceftazidime with AVE1330A, at a fixed concentration of 4 mg/L (MIC, ≤0.06/0.25 and 0.12/0.25, respectively).

Fixed ratio combinations of ceftazidime and AVE1330A at 4:1 and 4:2, respectively, demonstrated similar inhibitory activity against the β-lactamase producing isolates. However, MIC were generally 2–8 fold higher when compared to those achieved with ceftazadime plus a 4mg/L fixed concentration of AVE1330A.

Escherichia coli *tansformants expressing TEM, OXA, SHV and PSE β-lactamases (Table 2)*

AVE1330A was highly effective in protecting both aztreonam and ceftazidime against the hydrolytic activity of transformants of *Escherichia coli* expressing TEM 3, TEM 6, TEM 9, TEM 10 and OXA 5 enzymes which were active against aztreonam and ceftazidime when the two antimicrobials were tested alone.

Klebsiella *spp. expressing characterised extended spectrum β-lactamases (ESBL) (Table 3)*

In combination with AVE1330A, at a fixed concentration of 4 mg/L, both aztreonam and ceftazidime were protected from hydrolysis by a range of extended spectrum β-lactamases (ESBL) of the SHV-type.

Fixed ratio combinations of ceftazidime and AVE1330A at 4:1 and 4:2, respectively, demonstrated similar inhibitory activity against the β-lactamase producing isolates. However, MIC were generally 2-8 fold higher when compared to those achieved with ceftazadime plus a 4mg/L fixed concentration of AVE1330A.

Table 1. The comparative in vitro activity (mg/L) of ceftazidime and aztreonam + AVE1330A (at a fixed concentration of 4 mg/L), and ceftazidime in a 4:1 and 4:2 ratio, respectively, with AVE1330A against ceftazidime-resistant strains of species of the *Enterobacteriaceae*.

Species	Isolate ID	AMX	AMX-C (2:1)	AMX-C (4mg/L)	AMX-AVE (4mg/L)	FOX	AZT	AZT-AVE (4mg/L)	CAZ	CAZ-AVE (4mg/L)	CAZ-AVE 4:1	CAZ-AVE 4:2
E.coli	C16/3/08	>64	32	>64	4	>64	16	0.12	32	0.25	1	1
E.coli	C20/2/06	>64	16	32	≤0.06	32	64	≤0.06	16	0.5	2	1
E.coli	C20/3/06	>64	16	32	≤0.06	32	64	0.12	16	≤0.03	1	1
E.coli	C20/4/42	>64	16	32	≤0.06	32	64	≤0.06	16	0.25	2	1
E.coli	C22/1/24	>64	16	16	≤0.06	32	8	≤0.06	>128	1	8	4
E.coli	C23/2/08	>64	32	>64	2	16	4	≤0.06	32	0.25	2	1
E.coli	C23/3/22	>64	32	>64	1	32	4	≤0.06	128	0.5	2	2
E.coli	C3/2/21	>64	32	>64	16	>64	8	0.25	128	0.5	NT	NT
E.coli	C32/1/33	>64	16	32	≤0.06	16	64	≤0.06	32	0.25	0.5	0.5
E.coli	C32/3/24	>64	8	4	≤0.06	4	64	≤0.06	16	≤0.03	1	1
E.coli	C33/2/37	>64	16	64	2	4	16	≤0.06	16	0.25	0.25	0.25
E.coli	C4/2/39	>64	16	>64	≤0.06	64	>64	≤0.06	>128	≤0.03	16	8
C.freundii	C20/3/35	>64	32	>64	4	>64	8	0.12	128	0.25	1	1
C.freundii	C23/4/25	>64	32	>64	4	>64	64	0.25	128	0.5	2	2
C.freundii	C24/2/41	>64	32	>64	>64	64	64	0.12	>128	1	4	2
C.freundii	C34/4/09	>64	64	>64	4	>64	16	0.12	128	0.5	2	1
C.freundii	C4/4/06	>64	64	>64	16	>64	4	0.12	32	0.5	0.5	0.5
C.freundii	C7/2/44	>64	64	>64	4	>64	32	0.12	128	0.5	2	1
K.pneumoniae	C20/1/22	>64	8	8	4	4	>64	0.12	128	0.5	2	1
K.pneumoniae	C20/2/27	>64	4	2	≤0.06	2	64	≤0.06	128	≤0.03	1	1
K.pneumoniae	C20/2/32	>64	8	4	4	8	>64	0.25	>128	1	4	4
K.pneumoniae	C20/3/10	>64	16	64	16	4	>64	0.25	>128	1	4	4
K.pneumoniae	C24/2/20	>64	32	>64	32	8	2	0.12	128	1	4	4
K.pneumoniae	C4/2/11	>64	16	16	4	8	>64	0.5	>128	1	2	2
K.pneumoniae	C4/2/27	>64	16	16	4	8	>64	0.25	>128	1	4	4
K.pneumoniae	C4/2/28	>64	8	8	1	8	>64	0.12	128	0.5	2	2
K.pneumoniae	C4/2/30	>64	32	64	≤0.06	8	>64	0.12	>128	0.5	2	2
K.pneumoniae	C5/2/11	>64	16	32	≤0.06	16	>64	0.12	>128	0.5	4	2
K.pneumoniae	C5/2/39	>64	8	16	4	4	>64	0.12	128	0.5	2	2
K.oxytoca	C19/1/41	>64	16	16	16	4	>64	0.25	>128	4	4	4
E.cloacae	C18/4/15	>64	64	>64	64	>64	>64	1	>128	1	4	4
E.cloacae	C21/2/17	>64	64	>64	16	>64	32	0.25	128	0.5	2	2
E.cloacae	C22/2/30	>64	>64	>64	64	>64	64	1	128	2	4	4
E.cloacae	C24/2/43	>64	64	>64	16	>64	32	0.25	128	0.5	2	2
E.cloacae	C24/3/02	>64	64	>64	16	64	4	0.12	16	0.5	2	0.5
E.cloacae	C3/2/40	>64	64	>64	32	>64	32	0.5	128	1	4	2
E.cloacae	C31/2/42	>64	64	>64	16	>64	64	0.25	128	1	4	4
E.cloacae	C32/3/29	>64	64	>64	32	>64	0.5	≤0.06	16	0.25	4	2
E.cloacae	C32/4/36	>64	64	>64	>64	>64	64	0.5	128	8	8	8
E.cloacae	C33/4/13	>64	64	>64	32	>64	64	0.5	128	1	4	2
E.cloacae	C4/2/26	>64	32	>64	≤0.06	>64	>64	≤0.06	>128	0.5	4	4
E.cloacae	C7/4/44	>64	64	>64	32	>64	4	1	32	1	4	2
E.cloacae	C8/4/10	>64	64	>64	16	>64	32	0.5	128	0.5	2	2
M.morganii	C34/2/28	>64	>64	>64	8	16	2	≤0.06	16	0.12	0.5	0.25
M.morganii	C34/4/06	>64	>64	>64	32	16	8	0.25	64	0.25	2	0.5

Antimicrobial abbreviations: AMX = amoxycillin, AMX – C (2:1) = amoxycillin + clavulanic acid in a 2:1 ratio; **AMX-C** (4 mg/L) = amoxycillin + clavulanic acid at a fixed concentration of 4 mg/L, **AMX-AVE** = amoxycillin + AVE1330A at a fixed concentration of 4 mg/L; **FOX** = cefoxitin, **AZT**= aztreonam, **AZT-AVE** = aztreonam + AVE1330A at a fixed concentration of 4mg/L; **CAZ** = ceftazidime, **CAZ-AVE** = ceftazidime + AVE 1330A at a fixed concentration of 4mg/L; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:1; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:2

Table 2. The comparative in vitro activity (mg/L) of ceftazidime and aztreonam + AVE1330A (at a fixed concentration of 4 mg/L) against *Escherichia coli* transformants expressing various β-lactamase types.

Species	Plasmid type	AMX	AMX-C (2:1)	AMX-C (4mg/L)	AMX-AVE (4mg/L)	FOX	AZT	AZT-AVE (4mg/L)	CAZ	CAZ-AVE (4mg/L)	CAZ-AVE 4:1	CAZ-AVE 4:2
E.coli	TEM 1	>64	16	32	4	0.5	≤0.06	≤0.06	0.12	0.06	NT	NT
E.coli	TEM 1	>64	16	16	2	4	0.12	≤0.06	0.25	0.12	NT	NT
E.coli	TEM 2	>64	32	64	32	4	0.12	≤0.06	0.5	0.25	NT	NT
E.coli	TEM 3	>64	8	8	4	8	4	≤0.06	16	0.12	NT	NT
E.coli	TEM 6	>64	16	32	32	8	16	0.12	>128	0.5	NT	NT
E.coli	TEM 9	>64	16	16	16	4	64	,0.06	>128	0.5	NT	NT
E.coli	TEM 9	>64	16	16	16	4	64	≤0.06	>128	0.5	NT	NT
E.coli	TEM 10	>64	16	16	16	8	32	0.12	128	0.5	NT	NT
E.coli	OXA 1	>64	32	>64	32	8	0.12	0.12	0.25	0.25	NT	NT
E.coli	OXA 2	>64	16	32	32	4	≤0.06	≤0.06	0.25	0.12	NT	NT
E.coli	OXA 3	>64	16	16	16	4	0.12	0.12	0.5	0.25	NT	NT
E.coli	OXA 4	>64	32	64	32	4	0.12	≤0.06	0.25	0.25	NT	NT
E.coli	OXA 5	>64	8	8	8	4	16	0.12	64	0.5	NT	NT
E.coli	OXA 7	>64	32	>64	>64	4	2	0.5	0.5	0.25	NT	NT
E.coli	SHV 1	>64	8	8	2	4	0.12	≤0.06	0.25	0.12	NT	NT
E.coli	PSE 2	>64	32	64	>64	2	0.5	0.12	0.25	0.25	NT	NT
E.coli	PSE 2	16	4	4	4	0.5	≤0.06	≤0.06	0.12	0.06	NT	NT
E.coli	PSE 4	>64	16	16	>64	4	≤0.06	≤0.06	0.12	0.12	NT	NT

Antimicrobial abbreviations: AMX = amoxycillin, AMX – C (2:1) = amoxycillin + clavulanic acid in a 2:1 ratio; **AMX-C** (4 mg/L) = amoxycillin + clavulanic acid at a fixed concentration of 4 mg/L, **AMX-AVE** = amoxycillin + AVE1330A at a fixed concentration of 4 mg/L; **FOX** = cefoxitin, **AZT**= aztreonam, **AZT-AVE** = aztreonam + AVE1330A at a fixed concentration of 4mg/L; **CAZ** = ceftazidime, **CAZ-AVE** = ceftazidime + AVE 1330A at a fixed concentration of 4mg/L; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:1; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:2

Table 3. The comparative in vitro activity (mg/L) of ceftazidime and aztreonam + AVE 1330A (at a fixed concentration of 4 mg/L), and ceftazidime in a 4:1 and 4:2 ratio, respectively, with AVE1330A against *Klebsiella* spp. expressing characterised extended spectrum β-lactamases (ESBL).

Species	Isolate type	Plasmid	AMX (2:1)	AMX-C (4mg/L)	AMX-C (4mg/L)	AMX-AVE	FOX	AZT (4mg/L)	AZT-AVE	CAZ (4mg/L)	CAZ-AVE 4:1	CAZ-AVE 4:2	CAZ-AVE
K.pneumoniae	ESBL 813	SHV 2	>64	8	8	8	8	64	0.12	>64	0.5	2	1
Klebsiella sp	ESBL 152	SHV 2	>64	8	64	4	32	32	0.12	>64	0.5	1	1
Klebsiella sp	ESBL 633	SHV 2	>64	8	>64	8	16	16	≤0.06	>64	0.5	2	2
Klebsiella sp	ESBL 838	SHV 2	>64	>64	4	4	2	8	≤0.06	>64	0.12	2	2
Klebsiella sp	ESBL 1606	SHV 2	>64	>64	4	4	16	32	≤0.06	>64	0.12	2	1
Klebsiella sp	ESBL 1821	SHV 2	>64	32	>64	4	32	16	0.12	>64	0.5	2	2
Klebsiella sp	ESBL 1924	SHV 2	>64	32	>64	32	4	>64	1	>64	2	8	8
K.pneumoniae	ESBL 1964	SHV 3	>64	8	4	2	8	64	0.12	>64	0.5	2	2
Klebsiella sp	ESBL 606	SHV 3	>64	8	64	4	8	32	≤0.06	>64	0.25	4	2
Klebsiella sp	ESBL 963	SHV 3	>64	32	>64	8	4	8	≤0.06	>64	0.25	2	1
Klebsiella sp	ESBL 1953	SHV 3	>64	16	8	0.5	8	>64	≤0.06	>64	0.25	2	2
Klebsiella sp	ESBL 11	SHV 4	>64	4	2	2	32	64	0.12	64	0.25	1	1
Klebsiella sp	ESBL 256	SHV 4	>64	4	2	1	16	>64	≤0.06	>64	0.25	1	1
Klebsiella sp	ESBL 677	SHV 4	>64	4	8	1	8	64	≤0.06	>64	0.25	2	1
K.pneumoniae	ESBL 1851	SHV 5	>64	8	4	2	8	64	0.12	64	0.5	2	2
K.pneumoniae	ESBL 1972	SHV 5	>64	2	1	1	1	8	≤0.06	8	0.12	1	1
Klebsiella sp	ESBL 254	SHV 5	>64	8	32	1	4	>64	≤0.06	>64	0.12	2	1
Klebsiella sp	ESBL 679	SHV 5	>64	16	64	4	4	>64	≤0.06	>64	0.12	2	1
Klebsiella sp	ESBL 1453	SHV 5	>64	32	64	4	16	32	≤0.06	>64	0.25	2	1
Klebsiella sp	ESBL 1523	SHV 5	>64	32	64	8	4	32	≤0.06	>64	0.25	2	1
Klebsiella sp	ESBL 1664	SHV 5	>64	4	64	2	4	64	≤0.06	16	0.12	1	0.5
Klebsiella sp	ESBL 1970	SHV 2 & 5	>64	32	>64	4	8	4	≤0.06	16	0.06	2	1
K.pneumoniae	ESBL 961	SHV 3 & 2	>64	16	32	32	2	8	≤0.06	16	0.5	2	1
Klebsiella sp	ESBL 1960	SHV 3 & 5	>64	>64	4	0.5	2	>64	≤0.06	64	0.015	0.5	0.25

Antimicrobial abbreviations: AMX = amoxycillin, AMX – C (2:1) = amoxycillin + clavulanic acid in a 2:1 ratio; **AMX-C** (4 mg/L) = amoxycillin + clavulanic acid at a fixed concentration of 4 mg/L, **AMX-AVE** = amoxycillin + AVE1330A at a fixed concentration of 4 mg/L; **FOX** = cefoxitin, **AZT**= aztreonam, **AZT-AVE** = aztreonam + AVE1330A at a fixed concentration of 4mg/L; **CAZ** = ceftazidime, **CAZ-AVE** = ceftazidime + AVE 1330A at a fixed concentration of 4mg/L; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:1; **CAZ-AVE** = ceftazidime + AVE 1330A at a ratio of 4:2

CONCLUSIONS

- **AVE1330A is a potent inhibitor of a wide range of β-lactamases, including extended spectrum β-lactamases (ESBL).**
- **AVE1330A restores potent activity to both ceftazidime and aztreonam against ceftazidime-resistant isolates of species of the *Enterobacteriaceae.***
- **AVE1330A has clinical potential to extend the usefulness of either ceftazidime or aztreonam for the treatment of infection caused by β-lactam resistant strains of species of the *Enterobacteriaceae* when resistance is due to the expression of a wide range of β-lactamases, if its toxicological and pharmacokinetic profile is favourable.**

REFERENCES

1. Livermore, D. (1996). Are all β-lactams created equal? *Scan. J. Infect.* Dis. 101 (Suppl.), 33–43.
2. Vinokur, P.L., *et al.* (2001). Variations in the prevalence of strains expressing an extended-spectrum β-lactamase phenotype and characterisation of isolates from Europe, the Americas and the Western Pacific region. *Clin. Infect. Dis.* 32 (Suppl.2), S94–S103.
3. Pfaller, M.A. and Jones, R.N. (2002). Antimicrobial susceptibility of inducible AmpC β-lactamase producing Enterobacteriaceae from the Meropenem Yearly Susceptibility Test Information Collection (MYSTIC) programme, Europe 1997-2000. *Int. J. Antimicrob. Agents.* 19, 383–88.
4. Hall, G.H. and Barlow, M. (2004). Evolution of the serine β-lactamases: past, present and future. *Drug Resist. Updates.* 7(2), 111–123
5. Kaye, K.S. *et al.* (2004). Variety of β-lactamases produced by amoxycillin-clavulanate-resistant Esherichia coli isolated in the north-eastern United States. *Antimicrob. Agents Chemother.* 48(5), 1520–5
6. Bradford, P.A. (2001). Extended spectrum β-lactamases in the 21st century: characterisation, epidemiology and detection of this important resistance threat. *Clin. Micro. Rev.* 14, 933–51
7. Miller, L.A. *et al.* (2001). β-lactamase inhibitor combinations in the 21st century: current agents and new developments. *Curr. Opin. Pharmacol.* 1, 451–58
8. Bonnefoy, A. *et al.* (2004). In vitro activity of AVE1330A, an innovative broad-spectrum non-β-lactam β-lactamase inhibitor. *J. Antimicrob. Chemother.* 54, 410–417
9. NCCLS. (2003). Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard – Sixth Edition. *NCCLS document M7-A6 (ISBN 1-56238-486-4).* NCCLS, Wayne, Pennsylvania 19087–1898, USA. (NB. NCCLS re-named CLSI [Clinical Laboratory Standards Institute]).