In Vitro Activity of the New β-Lactamase Inhibitor NXL104 : Restoration of Ceftazidime (CAZ) Efficacy Against Carbapenem-Resistant *Enterobacteriaceae* Strains.

F1 - 318

C. MIOSSEC[1], L. POIREL[2], D. LIVERMORE[3], T. STACHYRA[1], P. NORDMANN[2], M. BLACK[1] and P. LEVASSEUR[1]

[1] Novexel, Romainville, France ; [2] Hopital de Bicêtre, South-Paris Medical School, France, [3] Health Protection Agency, London, UK

novel therapies for infectious disease

Contact Information:
christine.miossec@novexel.com

ABSTRACT

Background: NXL104 is a novel β-lactamase inhibitor able to inhibit both class A and class C β-lactamases *in vitro* and *in vivo*. CAZ is an oxyimino cephalosporin whose clinical utility is increasingly compromised by the spread of expanded-spectrum β-lactamases. We evaluated the *in vitro* efficacy of CAZ + NXL104 against strains with decreased susceptibility to carbapenems, either mediated by class A or D carbapenemases or by class A or C enzyme in combination with impermeability.

Methods: MIC values of CAZ, imipenem (IPM) and cefotaxime (CTX), alone and in combination with NXL104 at 4 μg/mL were determined by the CLSI agar dilution method; piperacillin/tazobactam was used as a reference antibiotic combination. IC_{50} values for enzyme inhibition were measured using crude cell extracts and nitrocefin.

Results: NXL104 effectively restored the activity of CAZ, CTX or IPM against isolates producing the class A carbapenemases: IMI-2, NMC-A, GES-2, -3, -4, KPC-2, and -3. CAZ/NXL104 activity against isolates with KPC-2 or –3 enzymes was remarkable, with MIC values of ≤0.015-0.5 μg/mL, as compared with 64->128 for CAZ alone.
The OXA-48 class D carbapenemase was also potently inhibited by NXL104 (IC_{50} value of 0.2 μM) and the activity of CAZ against isolates producing OXA-48 enzymes was fully restored by NXL104. In contrast, OXA-23, -40 and –58 (which mostly occur in *Acinetobacter* spp.) were only poorly inhibited with IC_{50} values in the low μM range.

Conclusions: NXL104 is the only inhibitor undergoing clinical development with a spectrum covering class A and class C β-lactamases; importantly, it potently inhibits a wide range of β-lactamases implicated in carbapenem resistance. CAZ/NXL104 combination could offer a useful alternative for treatment of infections due to carbapenem-resistant *Enterobacteriaceae* strains.

BACKGROUND

Carbapenems have the broadest antibacterial spectrum of all β-lactam antibiotics, and have been less compromised by resistance than any penicillin, cephalosporin or monobactam. This activity is mainly related to their stability to hydrolysis by most of the common class A, C and D enzymes; importantly, they can act as β-lactamases inhibitors by undergoing extremely slow hydrolysis by β-lactamases. Carbapenem antibiotics are usually regarded as reserve agents for the treatment of serious Gram-negative infections caused by multiresistant strains; the emergence of carbapenem-resistance is therefore worrysome as available alternative therapeutic options are restricted.

Acquired carbapenem resistance in *Enterobacteriaceae* is mediated by different mechanisms, production of β-lactamases being the most prevalent; other resistance mechanisms are conferred by an increased efflux of the β-lactam antibiotics, a decreased permeability of the outer membrane or by a combination of reduced permeability and high level production of a β-lactamase. Carbapenemases represent a heterogenous group of β-lactamases belonging to different classes : class A, and class C serine-enzymes, and class B metallo-enzymes. Carbapenem-hydrolysing β-lactamases include chromosomal, integron and plasmid encoded enzymes. The most important of the plasmid serine carbapenemases are the KPC β-lactamases in *Enterobacteriaceae* and the OXA-type carbapenemases in non-fermentive Gram-negatives.

NXL104 is a new non-β-lactam inhibitor of β-lactamases that displays a broad spectrum inhibition profile for both class A and class C enzymes; both types of enzymes are inactivated very efficiently at low IC_{50} values, with low turn-over numbers and long covalent intermediate half-lives (1). NXL104 has virtually no intrinsic antibacterial activity, but efficiently protects β-lactams from hydrolysis in a variety of class A and class C producing strains, including ESBL producers.

The emergence of carbapenem resistance in opportunistic *Enterobacteriaceae* isolates poses a serious threat to hospital medicine since many of these bacterial strains are resistant to almost all antibacterial agents. The aim of the study was to evaluate the antibacterial activity of ceftazidime / NXL104 combination against carbapenem-resistant strains.

REFERENCES

1. Bonnefoy A et al, J Antimicrob Chemother (2004), 54:410-417
2. Stachyra T et al, 47th ICAAC, Chicago, USA (2007), Abstract # F1-320
3. Mushtaq S et al, 47th ICAAC, Chicago, USA (2007), Abstract # F1-319

METHODS

Compounds
The following antimicrobials were used: ceftazidime pentahydrate (CAZ), piperacillin (PIP), imipenem (IPM), cefoxitin (CTX).
The following β-lactam inhibitors were used: clavulanate lithium (CLA), tazobactam (TAZ) and NXL104. PIP/TAZ combination, and NXL104 combinations were prepared at 4 mg/L fixed concentration of inhibitor.

FIGURE 1
Chemical structure of NXL104

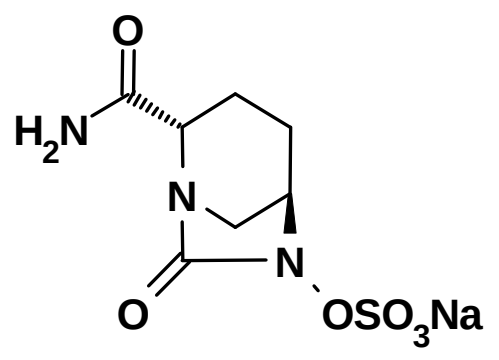
O
H_2N
N
N
O
OSO_3Na

Bacterial strains
Bacterial strains tested for susceptibility to cephalosporin / NXL104 combinations comprised:
- Five *E. coli* JM109 or DH5α transformants with sequenced carbapenemases
- Nine clinical *Enterobacteriaceae* (7 *Klebsiella spp*. and 2 *E. cloacae*) and one environmental strain (*E. asburiae*), all resistant to imipenem, expressing a type A, B or D carbapenemase.

Four *E. coli* JM109 transformants with an OXA enzyme were used to prepare cell extracts for determination of NXL104 inhibitory activity. These OXA-23, OXA-40, OXA-48 and OXA-58 β-lactamases represent the main sub-types of carbapenem-hydrolysing enzymes within the class D serine enzymes.

Susceptibility testing
MIC determination was performed using CLSI methods for antimicrobial susceptibility testing with Mueller-Hinton (MH) agar. MIC was defined as the lowest concentration which inhibited all visual growth.

β-lactamase assay
Crude bacterial extracts were prepared at 4℃ by vor texing ~$2x10^{10}$ bacterial cells with glass beads in 200 μL of 50 mM phosphate buffer, 2% glycerol, 0.1 mg/mL bovine serum albumin. Lysates were clarified by centrifugation and stored at –20℃.
β-lactamase activity was measured spectrophotometrically using nitrocefin as the reporter substrate.

Preliminary experiments were performed using serial dilutions of each cell lysate to define the optimal dilution for which the initial reaction rate (Vi) was constant over the 5 min of data collection. Then, in a second set of experiments, the inhibitory activity of NXL104, CLA and TAZ was determined by measuring IC_{50} values (11 serial dilutions of each inhibitor in the 100 μM to 2 nM range).
Inhibition of β-lactamase activity was determined at 37℃, after 30 min preincubation of enzyme and inhibitor. Reaction mixtures contained 20 μL of cell lysate at appropriate dilution, 50 μL of inhibitor, 20 μL of nitrocephin (100 μM final concentration), in a total volume of 200 μL of 50 mM pH 7.0 phosphate buffer.

RESULTS

TABLE 1
MIC values (μg/mL) of piperacillin / tazobactam (PIP/TAZ), imipenem (IPM) alone or in combination with NXL104, and cefatzidime (CAZ) alone or in combination with NXL104.
TAZ and NXL104 β-lactamase inhibitors were used at a fixed concentration (4 μg/mL).

Organism	Specimen ID	Mechanism / Class		Origin *	PIP-TAZ	IPM	IPM+ NXL104	Ratio IPM alone/ IPM+NXL104	CAZ	CAZ + NXL104	Ratio CAZ alone/ CAZ+NXL104
E. asburiae		IMI-2	A	E	2	> 512	8	>64	0.25	0.25	1
E. coli	JM109	NMC-A	A	L	2	1	nt	nt	0.25	≤0.015	≥16
E. cloacae		NMC-A	A	C	1	128	0.5	256	0.25	0.12	2
E. coli		GES-2	A	L	4	1	0.25	4	32	1	32
E. coli		GES-3	A	L	16	4	0.5	8	128	0.25	128
E. coli		GES-4	A	L	2	1	1	1	128	1	8
K. pneumoniae	YC	KPC-2	A	C	> 512	32	0.06	512	128	0.5	256
E. coli	DH5α	KPC-3	A	L	>128	8	nt	nt	128	0.25	512
K. pneumoniae	CL-5761	KPC-3	A	C	>128	>32	nt	nt	>128	≤0.015	>8192
K. pneumoniae	CL-5762A	KPC-3	A	C	>128	32	nt	nt	>128	≤0.015	>8192
K. pneumoniae	CL-5762B	KPC-3	A	C	>128	32	nt	nt	>128	≤0.015	>8192
K. pneumoniae	CL-5763	KPC-3	A	C	>128	>32	nt	nt	>128	≤0.015	>8192
K. pneumoniae		OXA-48 + SHV-2a	D	C	> 512	128	2	64	> 512	1	>512
Klebsiella spp.	Turkey kleb	OXA-48-like	D	C	>128	16	nt	nt	32	0.5	64
E. coli		VIM-1	B	L	256	8	8	1	>512	512	>1
E. coli		VIM-2	B	C	128	16	8	2	128	32	4

(*) : E = environmental strain ; C = clinical strain ; L = laboratory strain.

TABLE 2
IC_{50} values (μM) of NXL104, clavulanic acid and tazobactam against OXA carbapenemases (crude cell extracts ; mean values of at least 2 experiments)

	NXL104	CLA	TAZ
OXA-23	3.4	8.4	0.653
OXA-40	4.4	14.8	13.0
OXA-48	0.135	8.9	0.925
OXA-58	2.0	44.2	18.1

FIGURE 2
Inhibitory activity of NXL104, clavulanic acid and tazobactam against OXA-48 β-lactamase (crude cell extract)

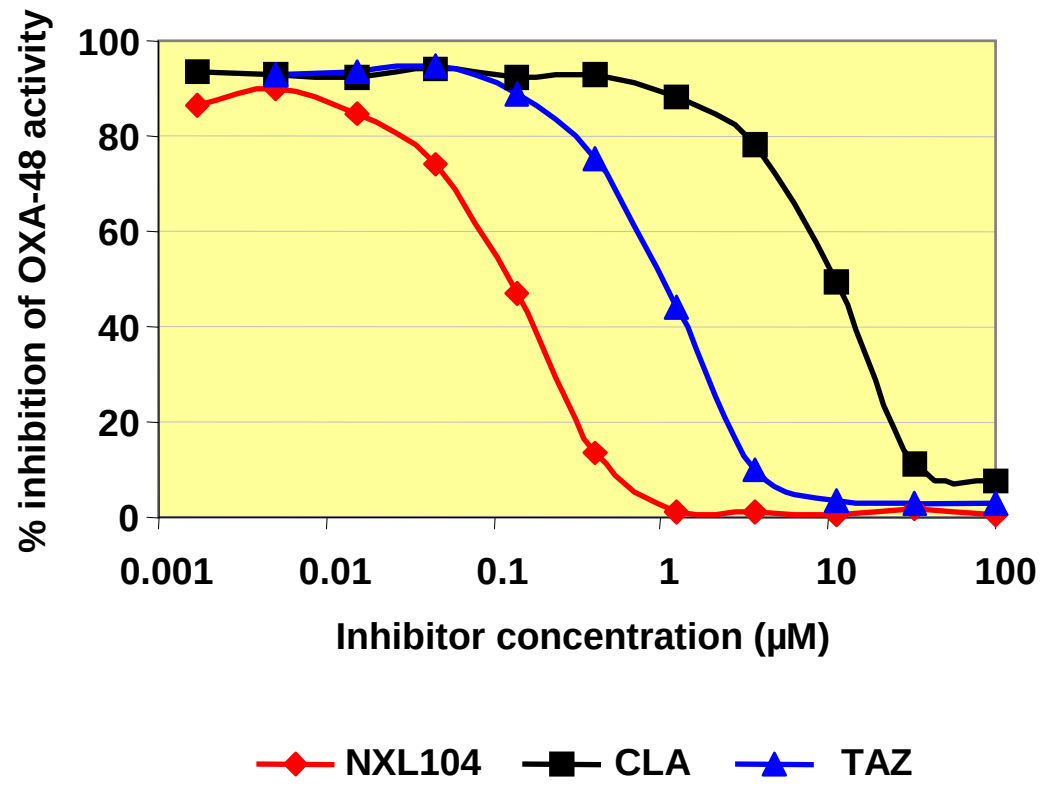
% inhibition of OXA-48 activity
100
80
60
40
20
0
0.001
0.01
0.1
1
10
100
Inhibitor concentration (μM)
NXL104
CLA
TAZ

CONCLUSIONS

The activity of NXL104 β-lactamase inhibtor against carbapenemases was evaluated by measuring the antibacterial activity of ceftazidime / NXL104 and imipenem / NXL104 combinations:

1 – Class A carbapenemases : NXL104 showed a potent activity against most class A carbapenemases (IMI-2, NMC-A, GES-2, -3, -4). In addition NXL104 was remarkably active against KPC-producing strains, as shown by ceftazidime MICs that were decreased by a >256 fold in the presence of the NXL104 inhibitor.

2 – Class B carbapenemases : NXL104 did not demonstrate any satisfactory activity against VIM metallo- β-lactamases.

3 – Class D carbapenemases : NXL104 potently restored the activities of ceftazidime and imipenem against OXA-48 producing isolates. The other carbapenem-hydrolysing OXAs were only marginally inhibited.

Overall, NXL104 demonstrated a clinically useful activity against Class A carbapenemases, including KPC enzymes (2). These plasmid-borne enzymes have a great potential to spread and KPC-producers cause extremely difficult to treat infections, due to their multidrug resistance. In addition, ceftazidime / NXL104 combination is known to be active against isolates for which carbapenem resistance is conferred by a combination of impermeability plus cephalosporinase production (3). The ceftazidime / NXL104 combination therefore offers a new option for treatment of carbapenem-resistant *Enterobacteriaceae* strains.

Presented at 47th ICAAC, Chicago, 2007